Effect of Chlorhexidine Gluconate on the Skin Integrity at PICC Line Sites

Marty Visscher, PhD[1], M. Victoria deCastro, MSN, RNC[2], Lisa Combs,RN[2], Lori Perkins, BSN, RN[2], Jill Winer[2], Nancy Schwegman, RNC[2], Claire Burkhart, BSN, RN[2], and Pattie Bondurant, MN, RN, CNS[2], Skin Sciences Institute[1], Regional Center for Newborn Intensive Care[2], Cincinnati Children's Hospital Medical Center, Cincinnati, OH

Introduction

The research literature on the effects of common skin care practices, e.g., cleansing, topical treatments, tapes, is sparse, particularly in high risk populations. NICU infants are at risk for skin breakdown due to prematurity, irritant exposure, stress, and adhesive tape removal. There is a need to minimize damage and facilitate the development of an effective stratum corneum (SC) barrier.

To reduce PICC line associated infections, the skin is treated with chlorhexidine gluconate (ChloraPrep®, 2% CHG, 70% alcohol, water) before insertion and application of tapes (steri-strips) and dressings (semipermeable, e.g., Tegaderm™). However, data on the skin effects, i.e., irritation, inflammation and SC barrier integrity, is limited. CHG (0.5%) was more effective than 10% povidone-iodine against colonization, but skin effects were not reported[1]. Severe contact dermatitis was seen in 5.7% of preterm infants treated with a CHG dressing (Biopatch®)[2]. We determined the skin effects of CHG at PICC sites among neonates in the Regional Center for Newborn Intensive Care (RCNIC) at Cincinnati Children's Hospital Medical Center (CCHMC).

References:

1. Linder N, et al. Acta Paedriatr 2004; 93:205-10.
2. Garland JS, et al. Pediatrics 2001; 107: 1431-6.
3. Visscher, et al. Arch Dermatol Res 2001; 293:491-499.

Hypothesis and Aims

Hypothesis:

- Treatment with CHG does not alter the normal skin barrier development in the high risk neonate, i.e., the condition of skin treated with CHG and a semipermeable dressing (Tegaderm™) will not differ from skin treated with the dressing alone (no CHG).

Aims:

In this research, we aim to:

- Evaluate the effects of chlorhexidine gluconate (Chloraprep®, 2% CHG, 70% alcohol, water) on the condition and barrier integrity of the skin at PICC line sites among infants in the neonatal intensive care unit.

Methods

Subjects:

- Gestational age: 23 – 39 wks
- PICC insertion site in arm or leg
- Expected to have a PICC for at least 18 days
- Parent/guardian provided written informed consent

Procedures:

- Compare three skin regions: (1) PICC site treated with CHG and a semipermeable dressing (Tegaderm™), (2) contralateral site treated with dressing alone (Tegaderm™, 2.5 cm2 piece) and (3) adjacent site with no treatment (control).
- Within subject design, subject is own control
- Skin evaluated at insertion and at weekly dressing changes
- Apply treatments at baseline (insertion) and weeks 1, 2, 3 to determine effects of repetitive exposure
- Measure skin irritation (erythema, dryness/scaling) and SC barrier function by transepidermal water loss (TEWL, $g/m^2/hr$) using the VapoMeter (Delfin Technologies, Inc.)
- Assess immediate irritant response to CHG application at PICC site

Measurements:

Score	Description	Area
0	None	---
0.5	Slight powderiness	< 10%
1.0	Slight powderiness or early cracking	10-50% < 10%
1.5	Slight powderiness or early cracking	>50% 10-50%
2.0	Early cracking or moderate cracking & scales	> 50% < 10%
2.5	Moderate cracking & scales	10-50%
3.0	Moderate cracking & scales	>50%
3.5	High cracking & lifting scales	10-50%
4.0	High cracking & lifting scales	>50%
4.5	Bleeding cracks	10-50%
5.0	Bleeding cracks	>50%

Skin Dryness Grading Scale

	Erythema	
0	None	
0.5	Faint – Def Pink	< 2%
1.0	Faint – Def Pink	2-10%
or	Definite Redness	< 2%
1.5	Faint – Def Pink	> 10%
or	Definite Redness	2-10%
or	Very intense redness	< 2%
2.0	Faint – Def Pink	> 50%
or	Definite Redness	10-50%
or	Very intense redness	2%
2.5	Definite Redness	> 50%
or	Very intense redness with edema	2-10%
3.0	Very intense redness with edema	> 10%

Skin Erythema Grading Scale

Statistical Analyses:

ANOVA for site comparisons of erythema, dryness/scaling, TEWL, $p < 0.05$; appropriate pairwise comparisons (SigmaStat, SPSS). Paired t-test for immediate erythema response. Linear mixed models (SPSS), repeated measures, F statistic at $p < 0.05$; treatment comparisons by method of Bonferroni.

Results

Subjects:

- 40 infants
- Gestational Age: 32.1 4.7 wks (range 23 – 39 wks)
- Age at Study Start: 34.8 5.5 wks (range 25 – 57 wks)
- Gender: 22 males, 18 females

Immediate Erythema:

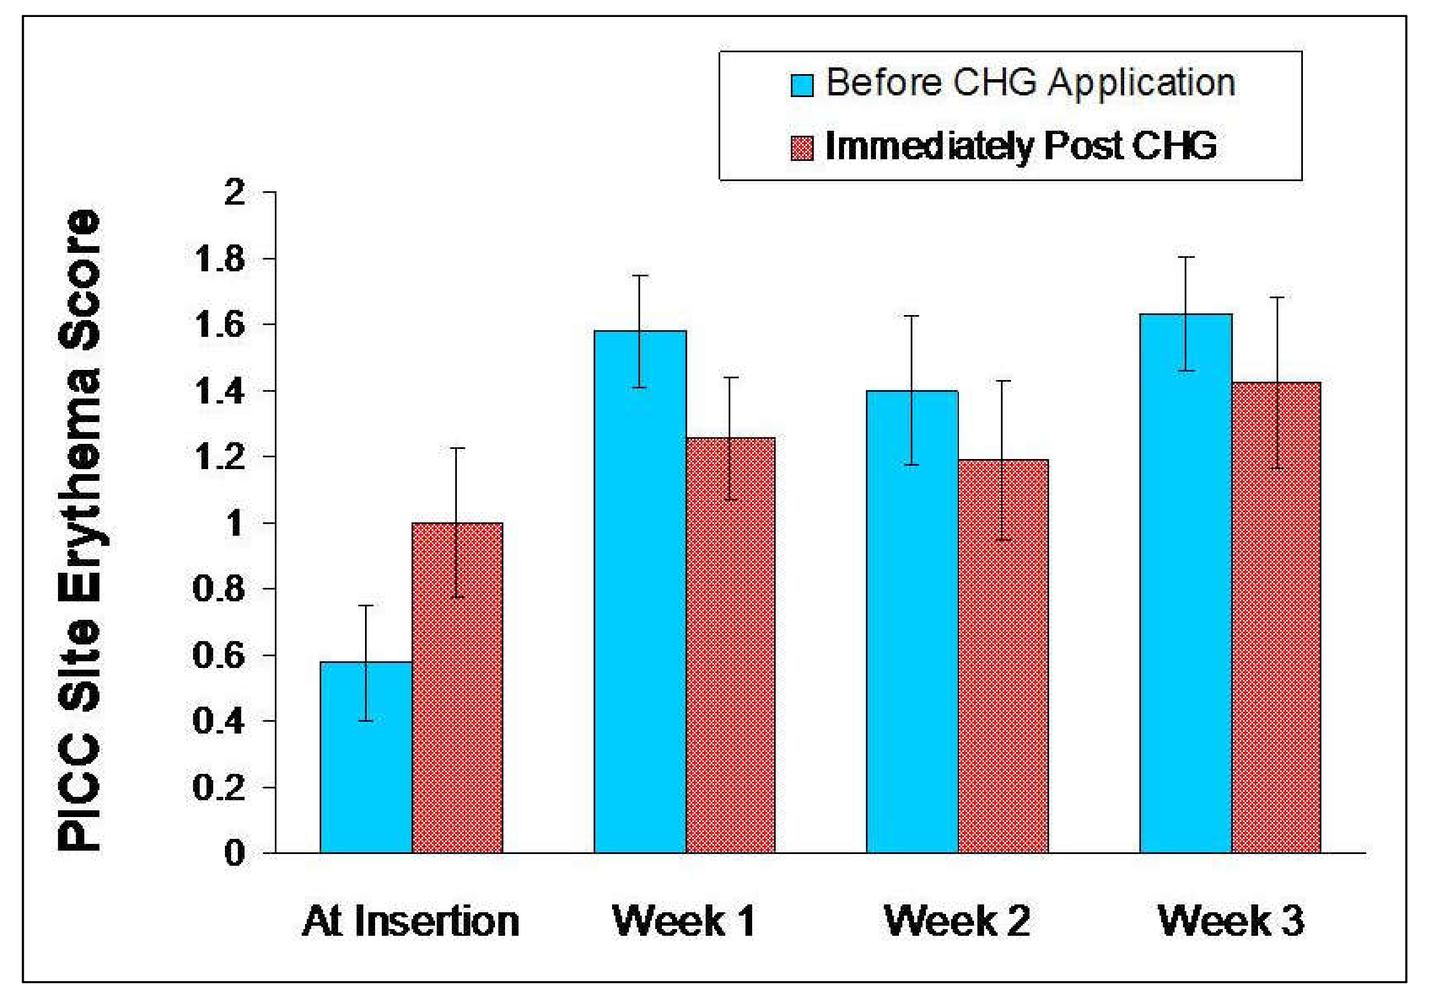
Before CHG Application
Immediately Post CHG
PICC Site Erythema Score
At Insertion
Week 1
Week 2
Week 3

Skin erythema immediately after CHG application (PICC insertion) was directionally increased (p = 0.07), but this effect was not observed for subsequent applications.

Erythema and Dryness:

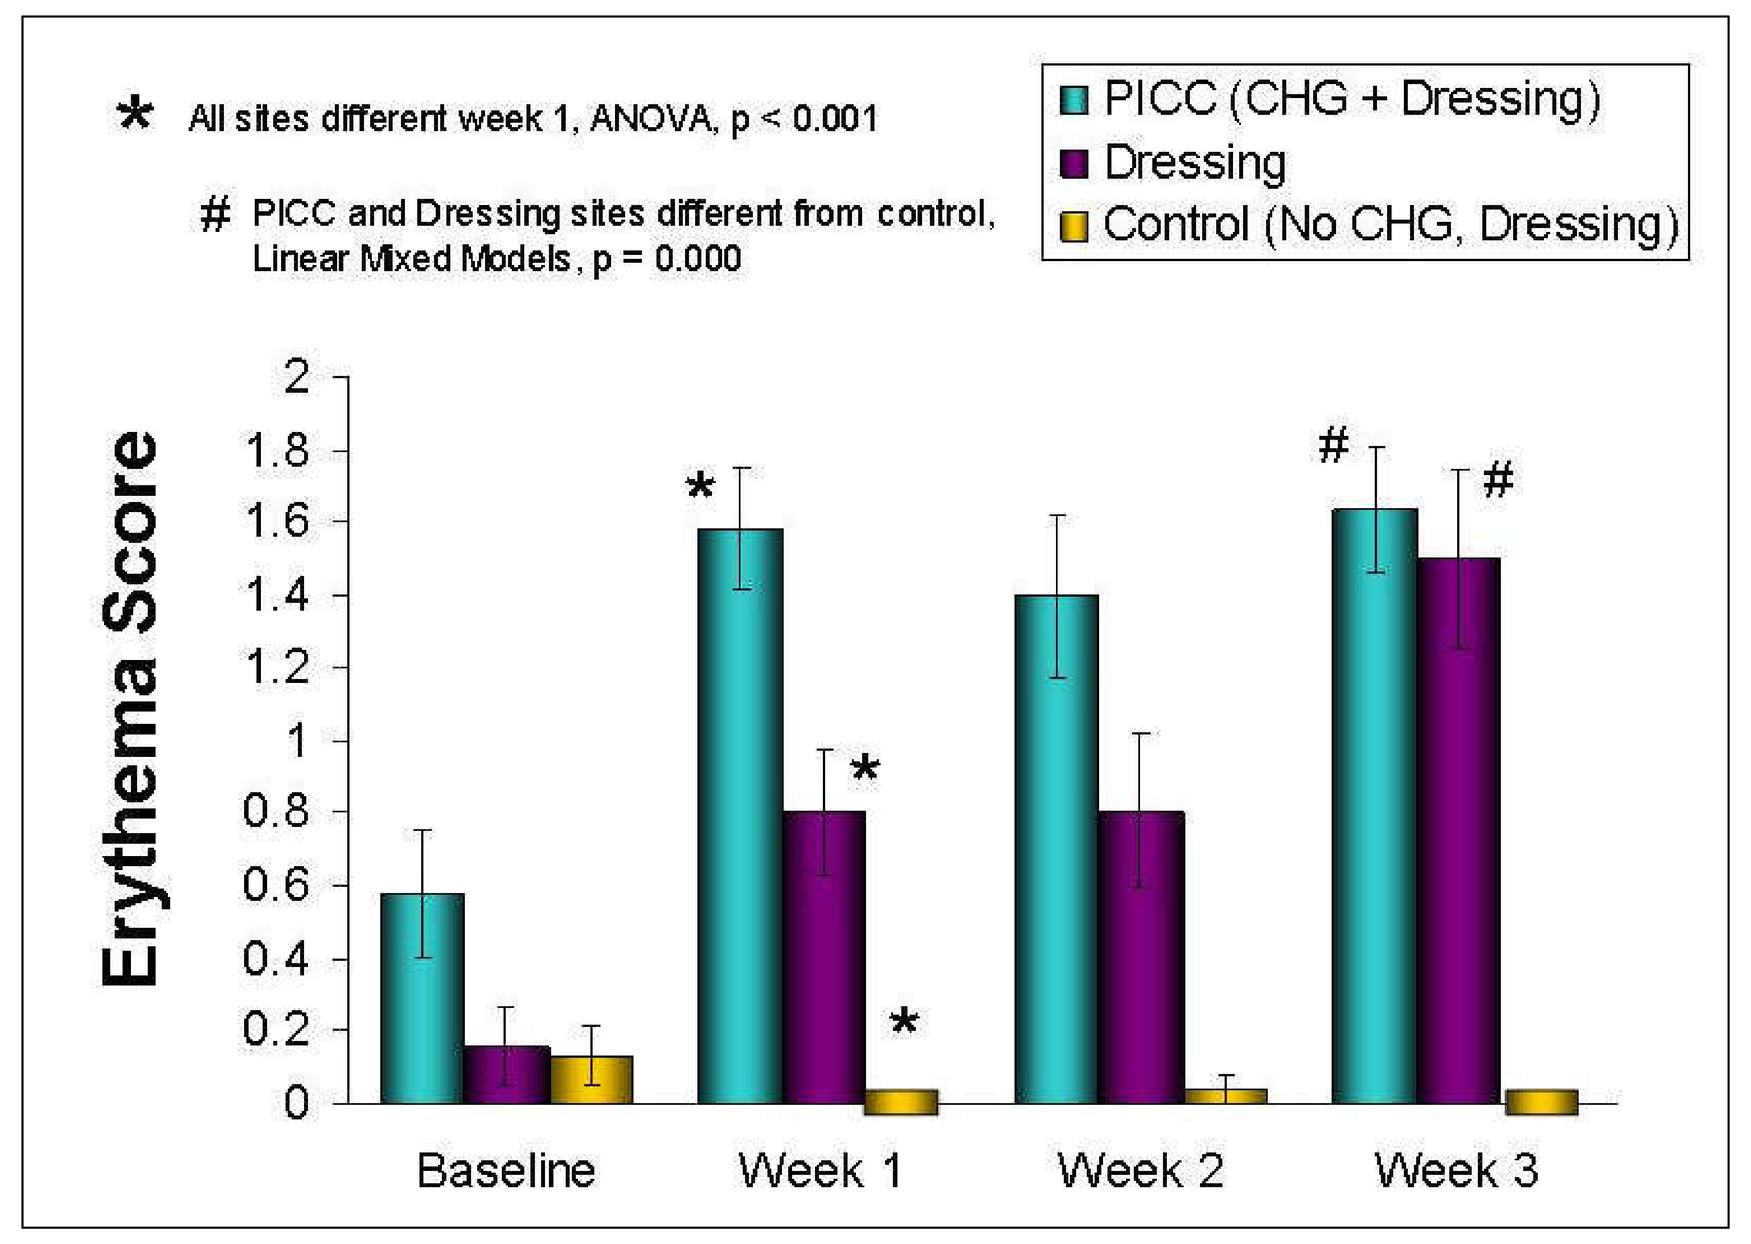
* All sites different week 1, ANOVA, p < 0.001
PICC and Dressing sites different from control, Linear Mixed Models, p = 0.000
PICC (CHG + Dressing)
Dressing
Control (No CHG, Dressing)
Erythema Score
Baseline
Week 1
Week 2
Week 3

After one week of exposure, the sites were significantly different for erythema (ANOVA, p < 0.001) with the highest score at the PICC site. By week 3, the PICC and dressing sites had comparable erythema and both were significantly higher than the control site.

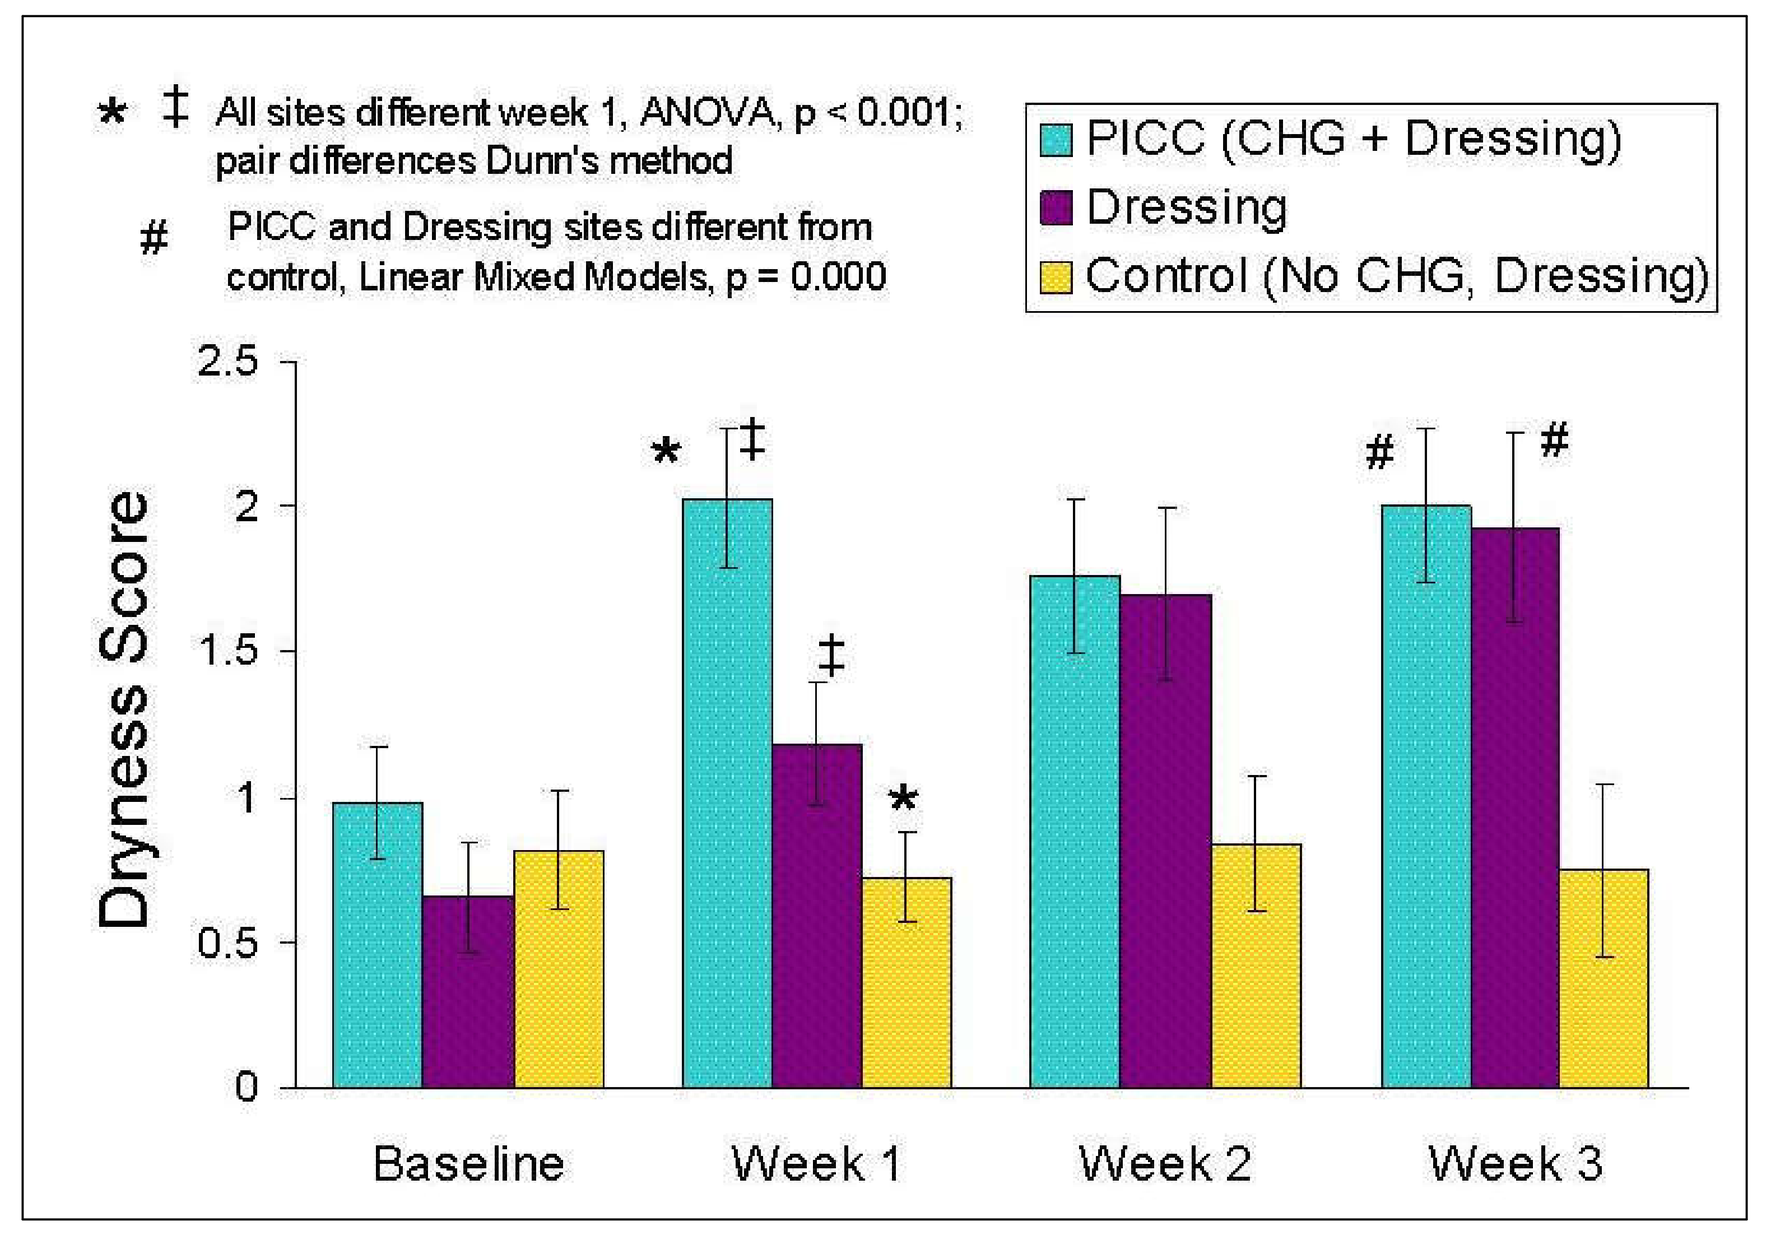
* ‡ All sites different week 1, ANOVA, p < 0.001; pair differences Dunn's method
PICC and Dressing sites different from control, Linear Mixed Models, p = 0.000
PICC (CHG + Dressing)
Dressing
Control (No CHG, Dressing)
Dryness Score
Baseline
Week 1
Week 2
Week 3

Dryness was observed at baseline. After one week of exposure, the PICC site had significantly higher skin dryness than the dressing site (ANOVA, p < 0.001). The PICC site was significantly drier than the control. By week 3, the PICC and dressing sites had comparable dryness and both were significantly higher than the control site.

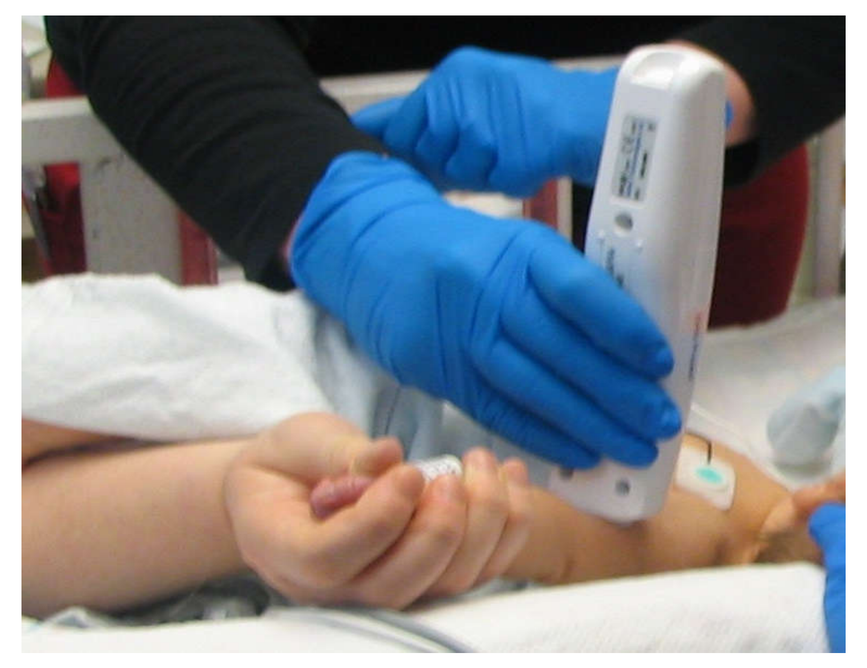

Measurement of TEWL (g.m2/hr) using the VapoMeter

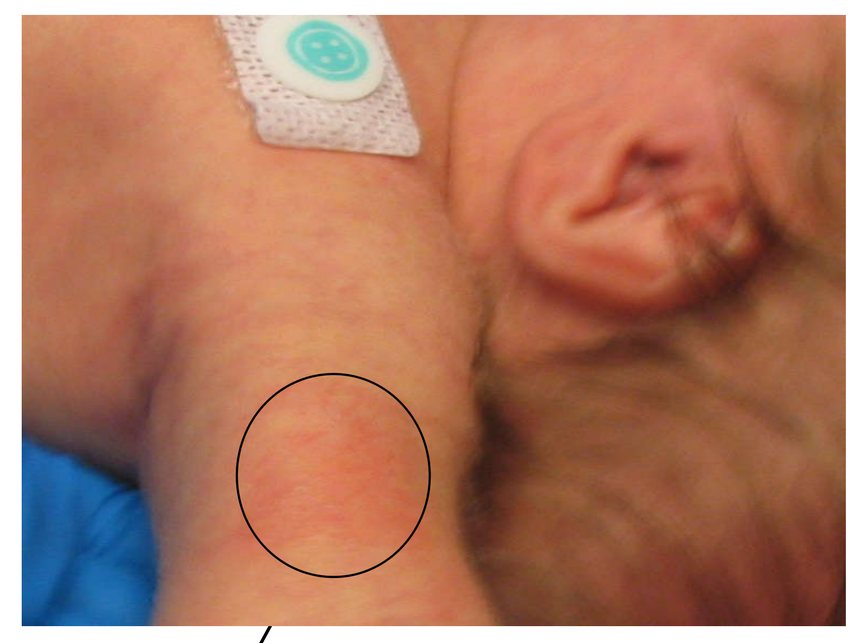

Skin erythema at a site treated with dressing alone is shown.

SC Barrier Integrity (TEWL):

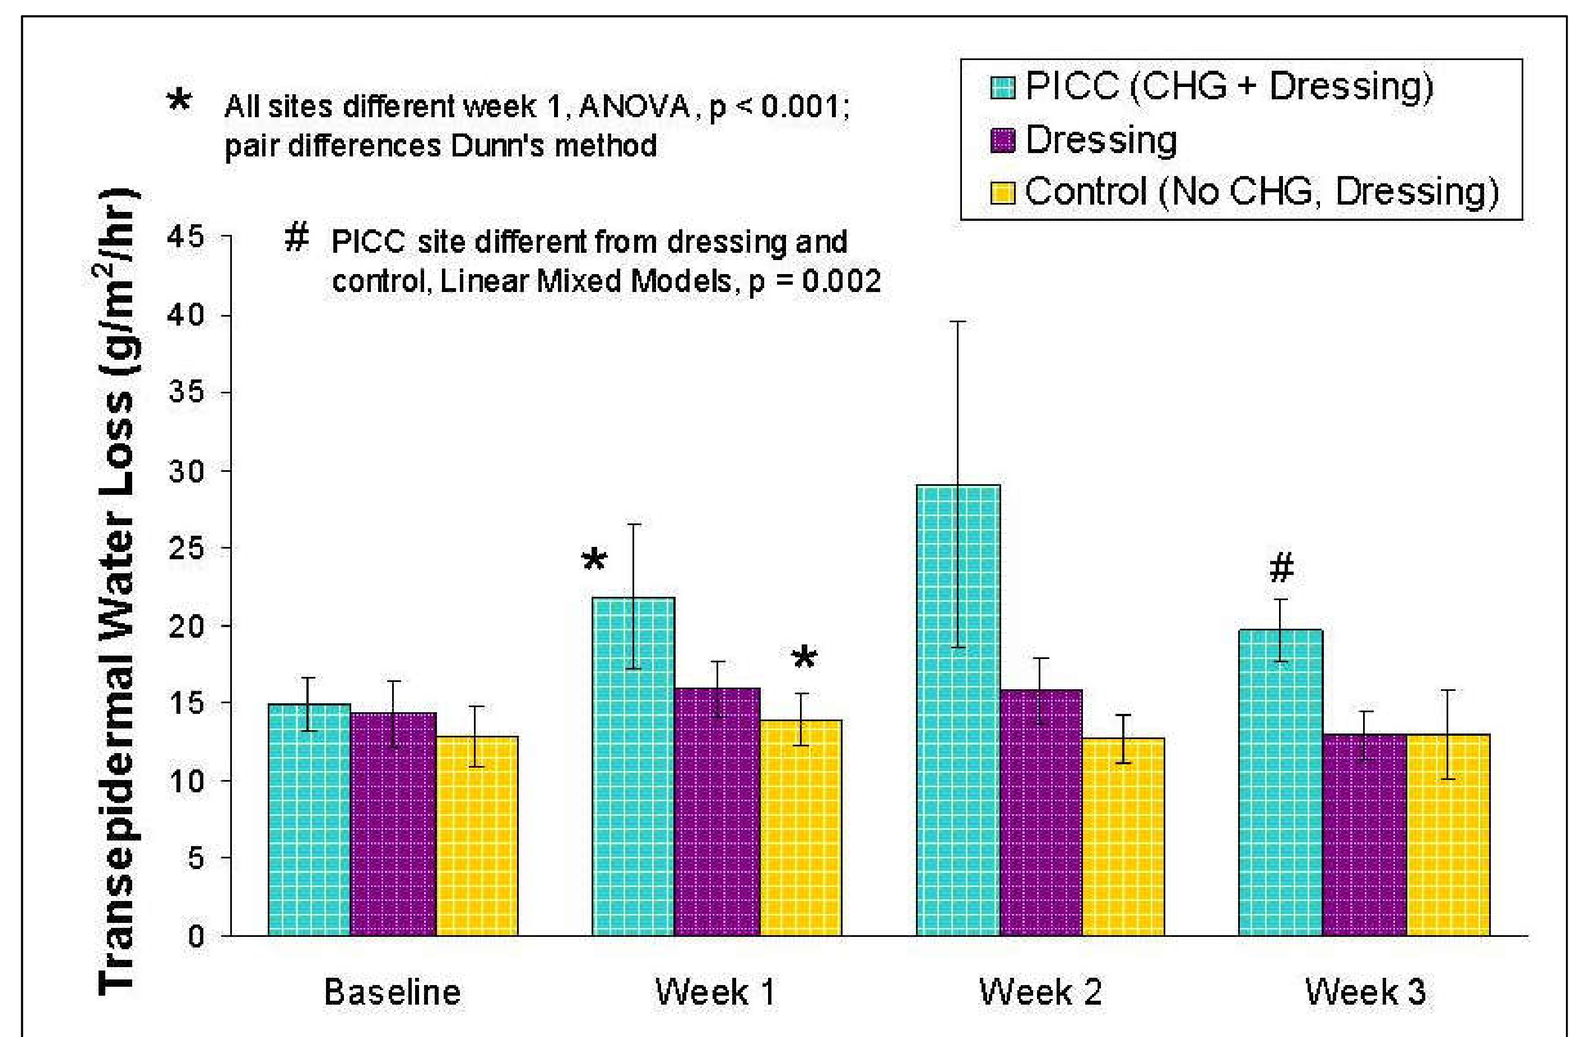
* All sites different week 1, ANOVA, p < 0.001; pair differences Dunn's method
PICC site different from dressing and control, Linear Mixed Models, p = 0.002
PICC (CHG + Dressing)
Dressing
Control (No CHG, Dressing)
Transepidermal Water Loss (g/m2/hr)
Baseline
Week 1
Week 2
Week 3

After one week, the PICC site had a significantly higher TEWL, indicating a compromised SC barrier, than the control. By week 3, TEWL at the PICC site was significantly higher (greater water loss) than both the dressing site and the control.

Conclusions and Implications

- CHG does not produce an immediate inflammatory response in this clinical population.
- The dressing (Tegaderm™) contributes to the PICC site erythema and dryness observed after prolonged exposure and repeated application.
- The combination of CHG + dressing may behave as a "low water vapor permeability" cover that allows water accumulation under it. Occlusion and water exposure delays skin barrier development and repair[3].
- The increase in TEWL at PICC sites may result from skin stripping at the time of dressing change.
- Dressings with inherently higher water vapor permeability are expected to minimize the skin breakdown at PICC sites. Investigation of alternatives is warranted.

Disclosure

An unrestricted educational grant was received from Enturia, Inc.